Characterizing Patients with Fibromyalgia: A US Real-World Claims Analysis

Poster #: RWD167

Errol Gould, PhD[1]; William Olsufka, PharmD[1]*

[1]Tonix Medicines, Inc., Berkeley Heights, NJ, USA; *Presenting author

BACKGROUND AND OBJECTIVES

- Fibromyalgia (FM) is a chronic, multisystem pain disorder characterized by widespread musculoskeletal pain, fatigue, sleep disturbance, cognitive dysfunction, and somatic symptoms, leading to substantial diagnostic and management challenges[1-4]
- In the United States, FM impacts approximately 10 million adults5 and is more common among women, who account for 75–90% of diagnosed cases[6]
- Patients with FM experience a high burden of comorbid conditions, including depression, anxiety, gastrointestinal disorders, migraine, and other chronic overlapping pain conditions, which exacerbate symptom severity, complicate treatment decisions, and increase clinical complexity[3,7-12]
- The combination of persistent symptoms, extensive comorbidities, and complex, multimodal treatment patterns contributes to substantial healthcare resource utilization (HCRU) and economic burden, with prior U.S. studies reporting wide-ranging annual per-patient healthcare costs driven largely by medications, outpatient visits, and inpatient services[8,13-18]
- The objective of this study was to analyze demographics, comorbidities, and healthcare resource utilization (HCRU) among US adults with FM using a large claims database

METHOD

- This retrospective cohort study utilized data from the Symphony Health closed claims database, encompassing administrative medical and pharmacy claims collected between April 2021 and April 2024, to evaluate real-world characteristics and HCRU among adults diagnosed with FM
- Adults aged ≥18 years with FM were identified using the ICD-10-CM diagnosis code M79.7, and patients were required to have continuous enrollment during the study period to ensure complete capture of healthcare encounters and utilization patterns
- The analyses focused on the third study year (2023–2024; Year 3 cohort), corresponding to the most recent and complete data available, to provide a contemporary assessment of disease burden in routine clinical practice
- Demographic characteristics, comorbidities, and HCRU were assessed descriptively, with comorbid conditions identified using ICD-10-CM diagnostic codes recorded in claims during the assessment period
- Medication use was assessed using the medication possession ratio (MPR), defined as the total days' supply of a medication divided by the length of the observation period, with values capped at 1.0 to account for early refills. For medication classes, MPR represents the aggregate duration of drug use during the year for which a patient had a supply of any drug within the class, providing an estimate of treatment exposure over time
- HCRU measures included office visits, pharmacy services, outpatient services, and inpatient care, with associated costs evaluated among patients with available Commercial or Medicare Advantage coverage

RESULTS

Figure 1. Patient characteristics, Year 3 cohort (N=261,776)

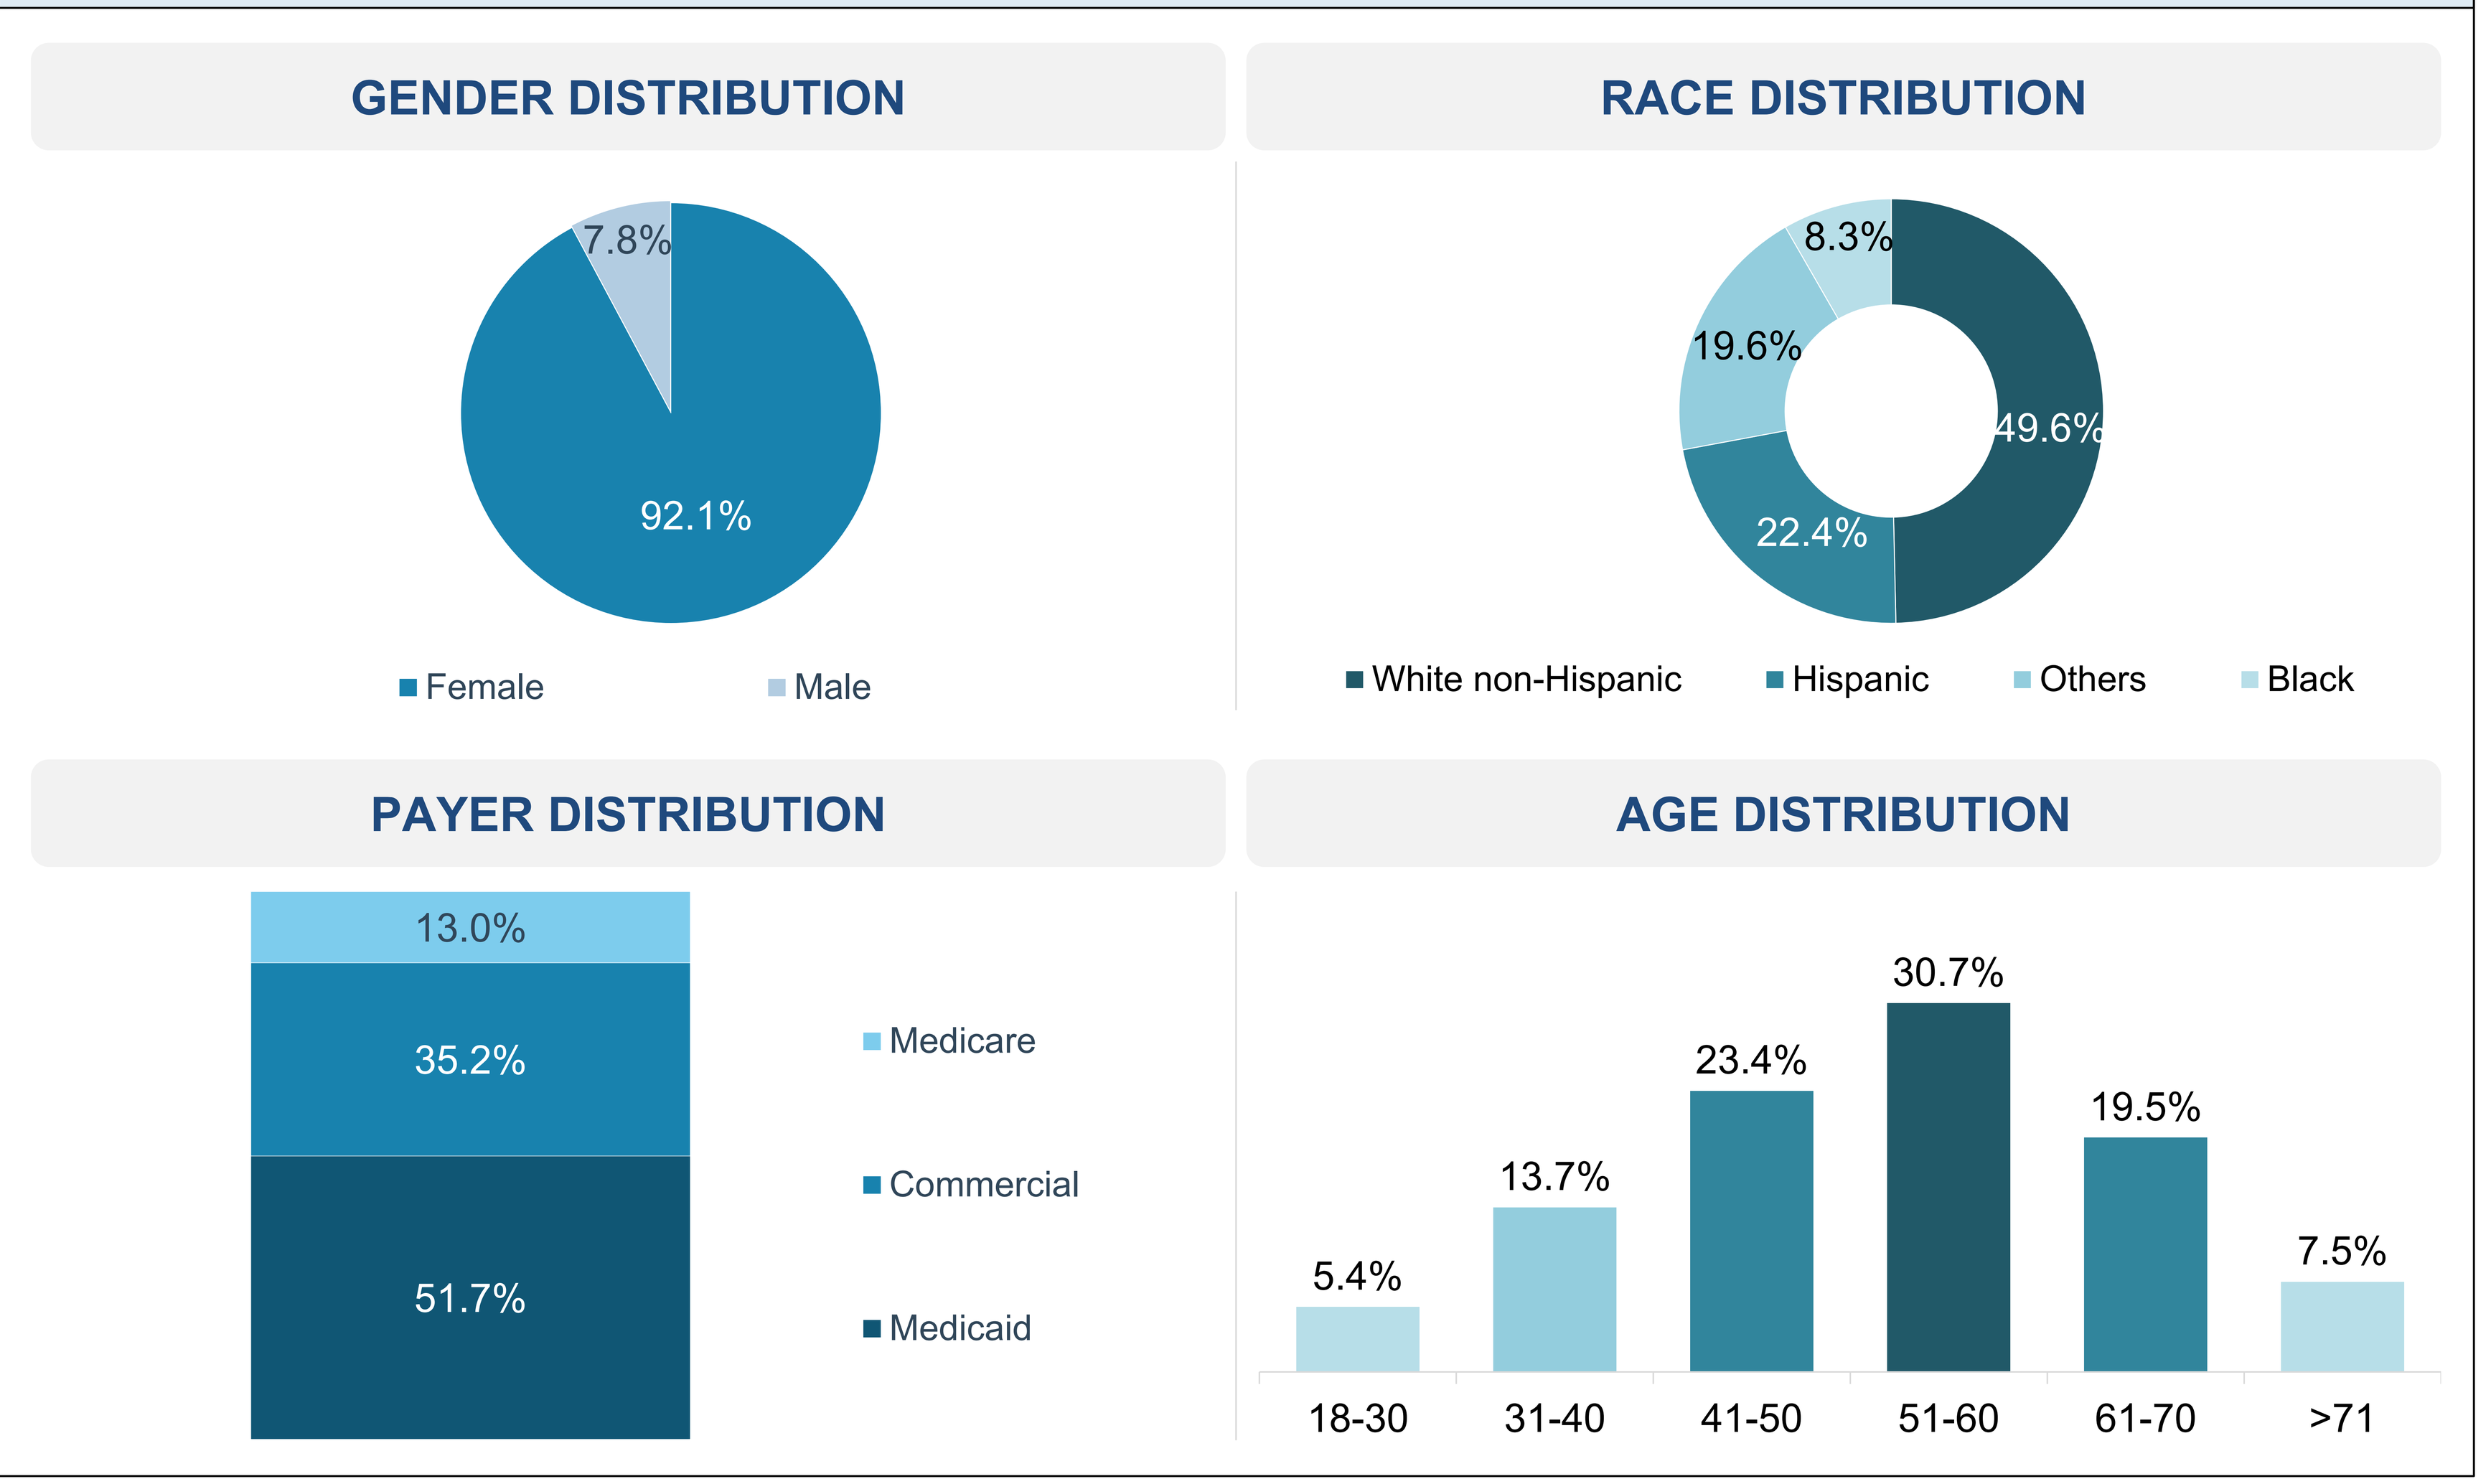
GENDER DISTRIBUTION
7.8%
92.1%
Female
Male
RACE DISTRIBUTION
8.3%
19.6%
49.6%
22.4%
White non-Hispanic
Hispanic
Others
Black
PAYER DISTRIBUTION
13.0%
35.2%
51.7%
Medicare
Commercial
Medicaid
AGE DISTRIBUTION
5.4%
13.7%
23.4%
30.7%
19.5%
7.5%
18-30
31-40
41-50
51-60
61-70
>71

RESULTS

- The Year 3 cohort included 261,776 adult patients diagnosed with FM, with a median age of 53 years (IQR: 43–61; range: 18–90; mean: 52.3 ± 13.0 years), reflecting a predominantly middle-aged population. Most patients were female (92.1%) and White non-Hispanic (49.6%) (Figure 1)
- Over half of the cohort was insured through Medicaid (51.7%), while 35.2% had Commercial insurance and 13.0% were covered by Medicare Advantage (Figure 1), indicating substantial representation of publicly insured patients
- Comorbidity burden was high, with 93.2% of patients having at least one FM-related comorbidity. The most prevalent comorbid conditions were anxiety/depression (63.9%), dorsalgia/back pain (56.5%), hypertensive diseases (51.4%), and joint pain (48.4%), highlighting the frequent coexistence of psychiatric and musculoskeletal conditions among patients with FM (Table 1)
- Most patients (90.3%; n=236,303) had at least one Elixhauser comorbidity. Among these patients, comorbidity burden was substantial, with 64.7% having 2 or more comorbid conditions. The distribution showed 19.2% with 2 comorbidities, 18.3% with 3 comorbidities, and 36.2% with 4 or more comorbidities, underscoring the substantial multimorbidity burden in the FM population (Table 2)
- Among patients with Commercial or Medicare Advantage coverage and at least one non-zero claim (n = 92,157), HCRU was extensive, with office visits reported for 93.6% of patients and pharmacy services utilized by 61.3%
- Inpatient services represented the largest component of healthcare costs, with mean annual inpatient costs of $29,896 per patient, followed by pharmacy costs ($9,453) and outpatient costs ($6,196) among insured patients. HCRU was extensive, with patients averaging 17.81 office visits, 26.74 pharmacy fills, 13.35 outpatient services, 2.95 ambulatory services, 2.58 inpatient encounters, and 2.12 emergency room visits annually (Figure 2)
- Antidepressants were the most frequently used medication class (48.2%), followed by NSAID analgesics (43.4%) and anticonvulsants (41.6%), with observed variations in drug use patterns across racial groups (Table 3)

Table 1. Comorbidities burden, Year 3 cohort (N=261,776)

	All patients	White women	Black/ Brown women	Women on opioids	Women with gut health issues	Older women with sleep issues
Number of patients	261,776 (100%)	119,486 (45.6%)	20,457 (7.8%)	95,514 (36.5%)	149,190 (57.0%)	68,319 (26.1%)
Any FM related comorbidities	93.2%	93.8%	94.6%	97.4%	96.8%	98.3%
Anxiety and depression	63.9%	69.2%	60.6%	72.1%	71.6%	76.5%
Dorsalgia (Back pain)	56.5%	56.3%	61.7%	70.0%	62.8%	65.7%
Joint pain	48.4%	47.9%	54.8%	59.5%	54.3%	58.0%
Hypertensive diseases	51.4%	49.2%	66.5%	59.8%	59.2%	68.9%
Osteoarthritis	36.4%	36.0%	43.4%	46.8%	43.1%	51.0%
Diabetes mellitus	26.5%	23.6%	33.9%	30.9%	31.4%	38.7%

Abbreviations: FM, fibromyalgia

- Adults with FM often experience a significant burden of comorbidities, with **over 90% of patients having at least one significant comorbidity**
- **Black and brown women tend to have a higher number of comorbidities** compared to white women
- **Women who are taking opioids or who have issues related to gut health or sleep tend to face a greater burden** than the overall FM population

Table 2. Elixhauser comorbidity counts across the FM cohort, Year 3 cohort (N=261,776)

Patients with number of Elixhauser comorbidities*, n (%)	Year 3 (04/23-03/24): n=261,776
With at least one comorbidity	236,303 (90.3)
1	43,267 (16.5)
2	50,314 (19.2)
3	48,006 (18.3)
4	38,690 (14.8)
5	26,984 (10.3)
>=6	29,042 (11.1)

*Top 10 Elixhauser comorbidities have been included in determining burden of comorbidity: 1. Hypertension; 2. Depression; 3. Obesity; 4. Chronic Pulmonary Disease; 5. Rheumatoid Arthritis/Collagen Disorders; 6. Diabetes; 7. Hypothyroidism; 8. Cardiac Arrhythmia; 9. Drug Abuse; 10. Fluid and Electrolyte Disorders.

Over one-third of patients (36.2%) had >3 comorbidities

Figure 2. Average annual service use per patient†, Year 3 cohort (Commercial / Medicare Advantage Patients With ≥1 Claim; N=92,157)

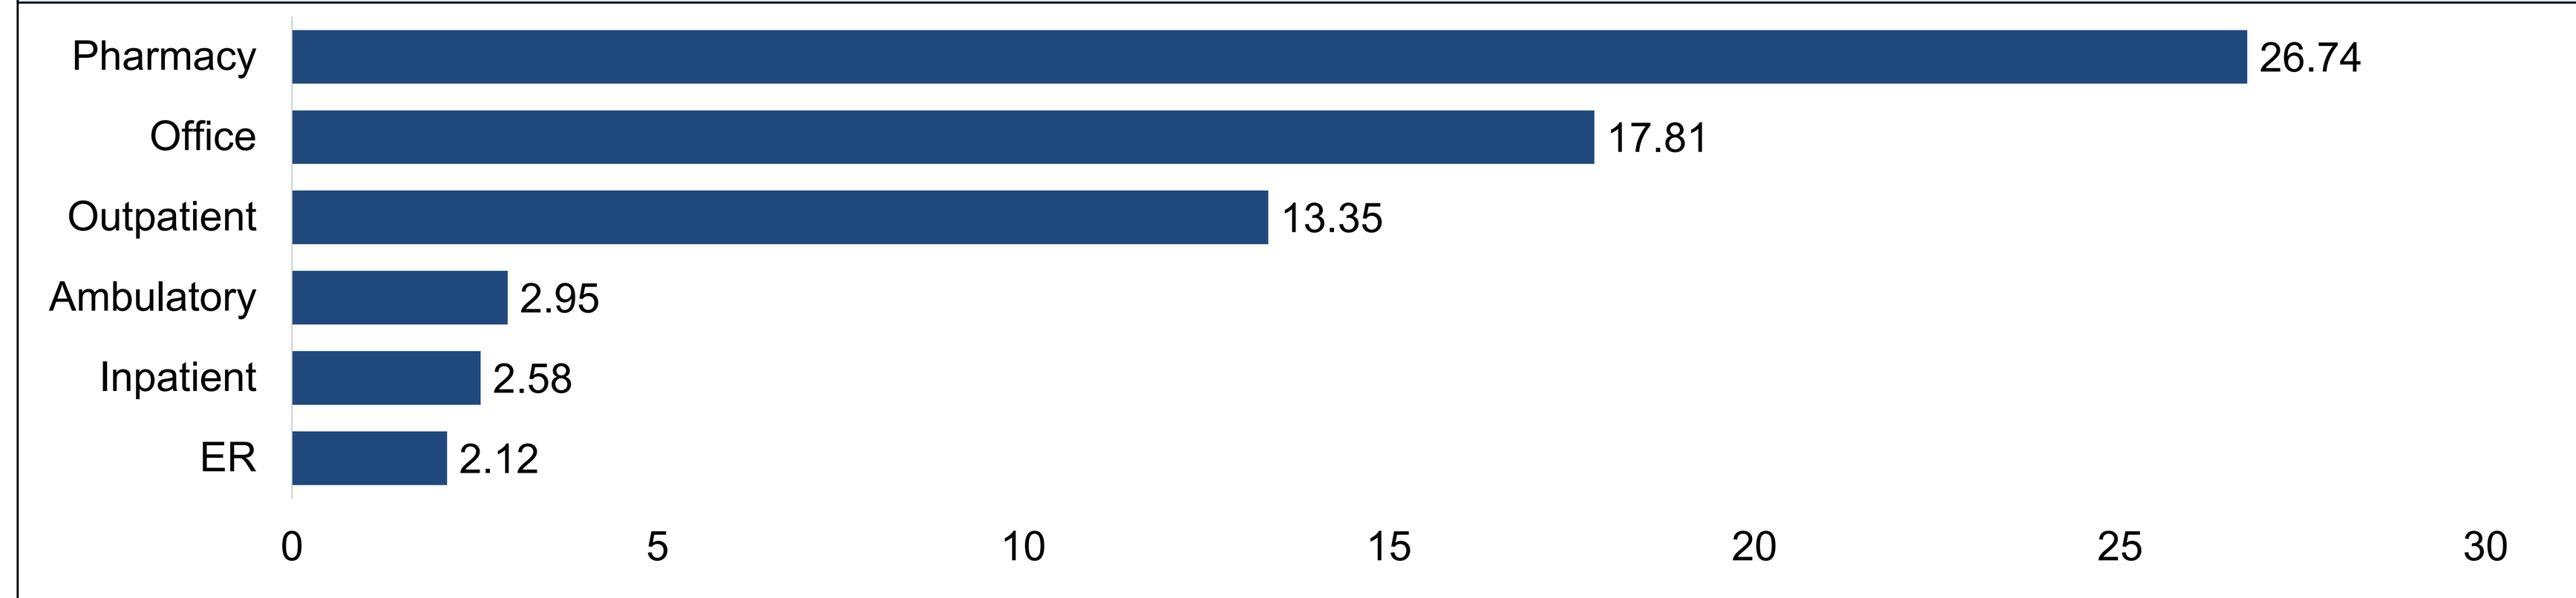
Pharmacy
Office
Outpatient
Ambulatory
Inpatient
ER
26.74
17.81
13.35
2.95
2.58
2.12
0
5
10
15
20
25
30

† Data includes only Commercial and Medicare Advantage patients. Patients diagnosed with FM in the previous year(s) have been assessed for the entire year; those diagnosed in the current year have been assessed from the date of first diagnosis in the year; Abbreviations: ER, emergency room.

Table 3. Drug use / poly-pharmacy, by drug class, Year 3 cohort (N=261,776)†

Percentage of Patients by Drug Class

	All patients	White women	Black/Brown women
Antidepressants	48.2	53.0	42.9
Analgesics - NSAID	43.4	41.4	48.4
Anticonvulsants	41.6	43.6	41.5
Gastric Acid Secretion Reducers	41.5	42.9	43.8
Skeletal Muscle Relaxants	40.7	42.2	45.9
Analgesics - Opioids	39.4	41.7	44.6

% of patients (scale: 40, 42, 44, 46, 48, 50, 52)

Medication Possession Ratio

	All patients	White women	Black/Brown women
Antidepressants	0.91	0.97	0.77
Analgesics – NSAID	0.34	0.35	0.30
Anticonvulsants	0.73	0.80	0.63
Gastric Acid Secretion Reducers	0.62	0.65	0.58
Skeletal Muscle Relaxants	0.39	0.42	0.35
Analgesics – Opioids	0.40	0.41	0.41

Medication Possession Ratio (MPR) (scale: 0.0, 0.2, 0.4, 0.6, 0.8, 1.0)

- **For antidepressants,** approximately **48%** of patients had a **high MPR** (around 0.9), suggesting that they were engaged in **long-term, consistent therapy** rather than short-term use
- Compared with White women, **Black/Brown women** show **higher use of pain- and gastrointestinal-related medications** (NSAIDs, opioids, muscle relaxants, gastric acid reducers), **but lower use and persistence (MPR) of centrally acting therapies** such as antidepressants and anticonvulsants

DISCUSSION

- These findings reinforce that FM is a multisystem condition associated with substantial clinical complexity, as evidenced by the high prevalence of comorbidities and extensive healthcare engagement observed in this large real-world population
- Medication use was widespread, with high utilization of antidepressants, analgesics, and anticonvulsants, reflecting reliance on multimodal pharmacologic strategies to manage pain, mood, and other FM-related symptoms in routine clinical practice
- Differences in treatment exposure and persistence, as measured by MPR, suggest variability in sustained use across drug classes, which may reflect differences in tolerability, effectiveness, or symptom targeting in real-world care
- The combination of high medication use, frequent healthcare encounters, and substantial inpatient costs highlights the ongoing clinical and economic burden of FM and underscores the need for more effective and durable management approaches

CONCLUSIONS

- FM affects mostly female, middle-aged patients.
- Adults with fibromyalgia experience a high clinical burden, with extensive comorbidities and widespread use of multimodal pharmacologic therapies
- HCRU and costs are substantial, driven largely by inpatient care and ongoing medication use, highlighting the need for comprehensive disease management and more effective therapeutic interventions to reduce the clinical and economic burden

LIMITATIONS

- Analyses relied on administrative claims data, which do not capture clinical severity, symptom burden, or confirmed diagnoses and may be affected by coding inaccuracies or misclassification
- Diagnoses, treatments, and outcomes were inferred from billing records rather than clinical assessment, which may result in under- or over-representation of certain conditions or events

REFERENCES

1.Bilge U, Sari YE, Balcioglu H, Bilge NSY, Kasifoglu T, Kayhan M, et al. Prevalence of comorbid diseases in patients with fibromyalgia: A retrospective cross-sectional study. Cardiovasc Dis. 2018;155:30–5
2.Choy EH, Mease PJ. Key symptom domains to be assessed in fibromyalgia (outcome measures in rheumatoid arthritis clinical trials). Rheum Dis Clin North Am. 2009;35(2):329–37
3.Rivera FA, Munipalli B, Allman ME, Hodge DO, Wieczorek MA, Wang B, et al. A retrospective analysis of the prevalence and impact of associated comorbidities on fibromyalgia outcomes in a tertiary care center. Front Med (Lausanne). 2023;10:1301944
4.Winslow BT, Vandal C, Dang L. Fibromyalgia: Diagnosis and Management. Am Fam Physician. 2023;107(2):137–44.
5.Fibromyalgia prevalence. National Fibromyalgia Association. Accessed July 3, 2025. https://www.fmaware.org/fibromyalgia-prevalence/.
6.Association NF. About Fibromyalgia 2025 [updated 2025/12/05. Available from: https://www.fmaware.org/about-fibromyalgia/.
7.Sedda S, Cadoni MPL, Medici S, Aiello E, Erre GL, Nivoli AM, et al. Fibromyalgia, depression, and autoimmune disorders: An interconnected web of inflammation. Biomedicines. 2025;13(2):503
8.D'Onghia M, Ciaffi J, Ruscitti P, Cipriani P, Giacomelli R, Ablin JN, et al. The economic burden of fibromyalgia: A systematic literature review. Semin Arthritis Rheum. 2022;56:152060
9.Erdrich S, Hawrelak JA, Myers SP, Harnett JE. A systematic review of the association between fibromyalgia and functional gastrointestinal disorders. Therap Adv Gastroenterol. 2020;13:1756284820977402
10.Tarar ZI, Farooq U, Nawaz A, Gandhi M, Ghouri YA, Bhatt A, et al. Prevalence of fibromyalgia and chronic fatigue syndrome among individuals with irritable bowel syndrome: An analysis of the United States National Inpatient Sample Database. Biomedicines. 2023;11(10):2594
11.Bothelius K, Kosek E, Jansson-Fröjmark M. Associations between sleep disturbance and functional impairment in fibromyalgia: A path analysis study. J Sleep Res. 2025;34:e70199
12.Lima D, Pacheco-Barrios K, Slawka E, Camargo L, Castelo-Branco L, Cardenas-Rojas A, et al. The role of symptom severity, heart rate, and central sensitization in predicting sleep quality in fibromyalgia. Pain Med. 2023;24(10):1153–60
13.Boonen A, van den Heuvel R, van Tubergen A, Goossens M, Severens JL, van der Heijde D, et al. Large differences in cost of illness and wellbeing between patients with fibromyalgia, chronic low back pain, or ankylosing spondylitis. Ann Rheum Dis. 2005;64(3):396–402
14.Knight T, Schaefer C, Chandran A, Zlateva G, Winkelmann A, Perrot S. Health-resource use and costs associated with fibromyalgia in France, Germany, and the United States. ClinicoEconomics Outcomes Res. 2013:171–80
15.Lacasse A, Bourgault P, Choinière M. Fibromyalgia-related costs and loss of productivity: A substantial societal burden. BMC Musculoskelet Disord. 2016;17(1):168
16.Perrot S, Schaefer C, Knight T, Hufstader M, Chandran AB, Zlateva G. Societal and individual burden of illness among fibromyalgia patients in France: Association between disease severity and OMERACT core domains. BMC Musculoskelet Disord. 2012;13:22
17.Penrod JR, Bernatsky S, Adam V, Baron M, Dayan N, Dobkin PL. Health services costs and their determinants in women with fibromyalgia. J Rheumatol. 2004;31(7):1391
18.Thompson JM, Luedtke CA, Oh TH, Shah ND, Long KH, King S, et al. Direct medical costs in patients with fibromyalgia: Cost of illness and impact of a brief multidisciplinary treatment program. Am J Phys Med Rehabil. 2011;90(1):40–6.

DISCLOSURES

EG, WO: Employee of Tonix Medicines, Inc.; holds stock and/or stock options in Tonix Pharmaceuticals Holding Corp

SPONSORSHIP

Tonix Medicines, Inc.

CONTACT INFO

errol.gould@tonixpharma.com; william.olsufka@tonixpharma.com