TNX-102 SL, Cyclobenzaprine HCl Sublingual Tablets, Demonstrates Pain Reduction and Favorable Tolerability in Participants With Fibromyalgia

Poster #37

Gregory M. Sullivan, MD[1]; Errol Gould, PhD[2]; Mary Kelley, MPH[1]; Jean Engels, MS[1]; Seth Lederman, MD[1]

[1]Tonix Pharmaceuticals, Inc., Chatham, NJ, USA; [2]Tonix Medicines, Inc., Chatham, NJ, USA

INTRODUCTION

- Fibromyalgia (FM) is a chronic condition characterized by widespread musculoskeletal pain, fatigue, sleep disturbances, and cognitive and somatic complaints[1,2]
 - FM has recently been described as the prototypic nociplastic syndrome[3]
 - Nociplastic pain is distinct from nociceptive or neuropathic pain and is characterized by pain arising from altered nociception in the absence of peripheral nociceptor or somatosensory system pathology[3,4]
 - Disruptions to the deep, restorative stages of sleep occur in patients with FM[5]
 - Poor sleep quality is highly associated with an amplification of nociplastic pain[4]
- In the US, FM affects an estimated 2%-8% of adults, 75%-90% of whom are female[6,7]
- FDA-approved treatments for FM have historically been limited by intolerable side effects that often lead to poor adherence[8]
- Tonmya™ (TNX-102 SL, cyclobenzaprine HCl sublingual tablets) is FDA approved for the treatment of FM in adults[9-11]
 - TNX-102 SL treatment has shown statistically significant reductions in daily pain in two pivotal phase 3 studies, RELIEF (NCT04172831) and RESILIENT (NCT05273749)
 - TNX-102 SL also improved sleep quality, fatigue, and cognitive function and was well tolerated, with a favorable safety profile
 - The demonstrated efficacy and tolerability of TNX-102 SL may support its addition to the existing therapeutic landscape

OBJECTIVE

- To report pain, sexual function, and tolerability outcomes for TNX-102 SL in the treatment of FM from RESILIENT, a phase 3, randomized, placebo-controlled study

METHODS

- RESILIENT was a 14-week, randomized, parallel-group, double-blind, placebo-controlled trial conducted at 34 sites in the US and was designed to evaluate the efficacy and safety of TNX-102 SL in participants with FM
 - Participants 18-65 years of age were randomized 1:1 to receive 2.8 mg TNX-102 SL (1 tablet) sublingually at bedtime for 2 weeks followed by 5.6 mg (2 tablets) for 12 weeks or matching placebo for 14 weeks
- The prespecified primary endpoint was change from baseline to Week 14 in the weekly average of daily self-reported average pain intensity scores (11-point numerical rating scale [NRS]), analyzed using a mixed model for repeated measures (MMRM) with multiple imputation for missing data
- Exploratory efficacy endpoints included changes in sexual function, as measured by the Changes in Sexual Functioning Questionnaire short form (CSFQ-14)
 - The CSFQ-14 was analyzed by ANCOVA, and significance testing was based on least-squares (LS) means using a 2-sided α=0.05 test. Two-sided 95% confidence intervals (CIs) were generated.
- Safety evaluations included changes in blood pressure, adverse events (AEs), other vital signs, weight, physical exams, Columbia-Suicide Severity Rating Scale (C-SSRS), and Beck Depression Inventory (BDI-II)

RESULTS

Participants

- A total of 813 participants were screened for eligibility, with 456 in the intent-to-treat (ITT) population receiving TNX-102 SL (n=231) or placebo (n=225) (**Figure 1**)
 - 81.0% of the TNX-102 SL group and 79.2% of the placebo group completed the study
- Participant demographics and baseline characteristics were generally comparable between groups (**Table 1**)

Figure 1: CONSORT Diagram

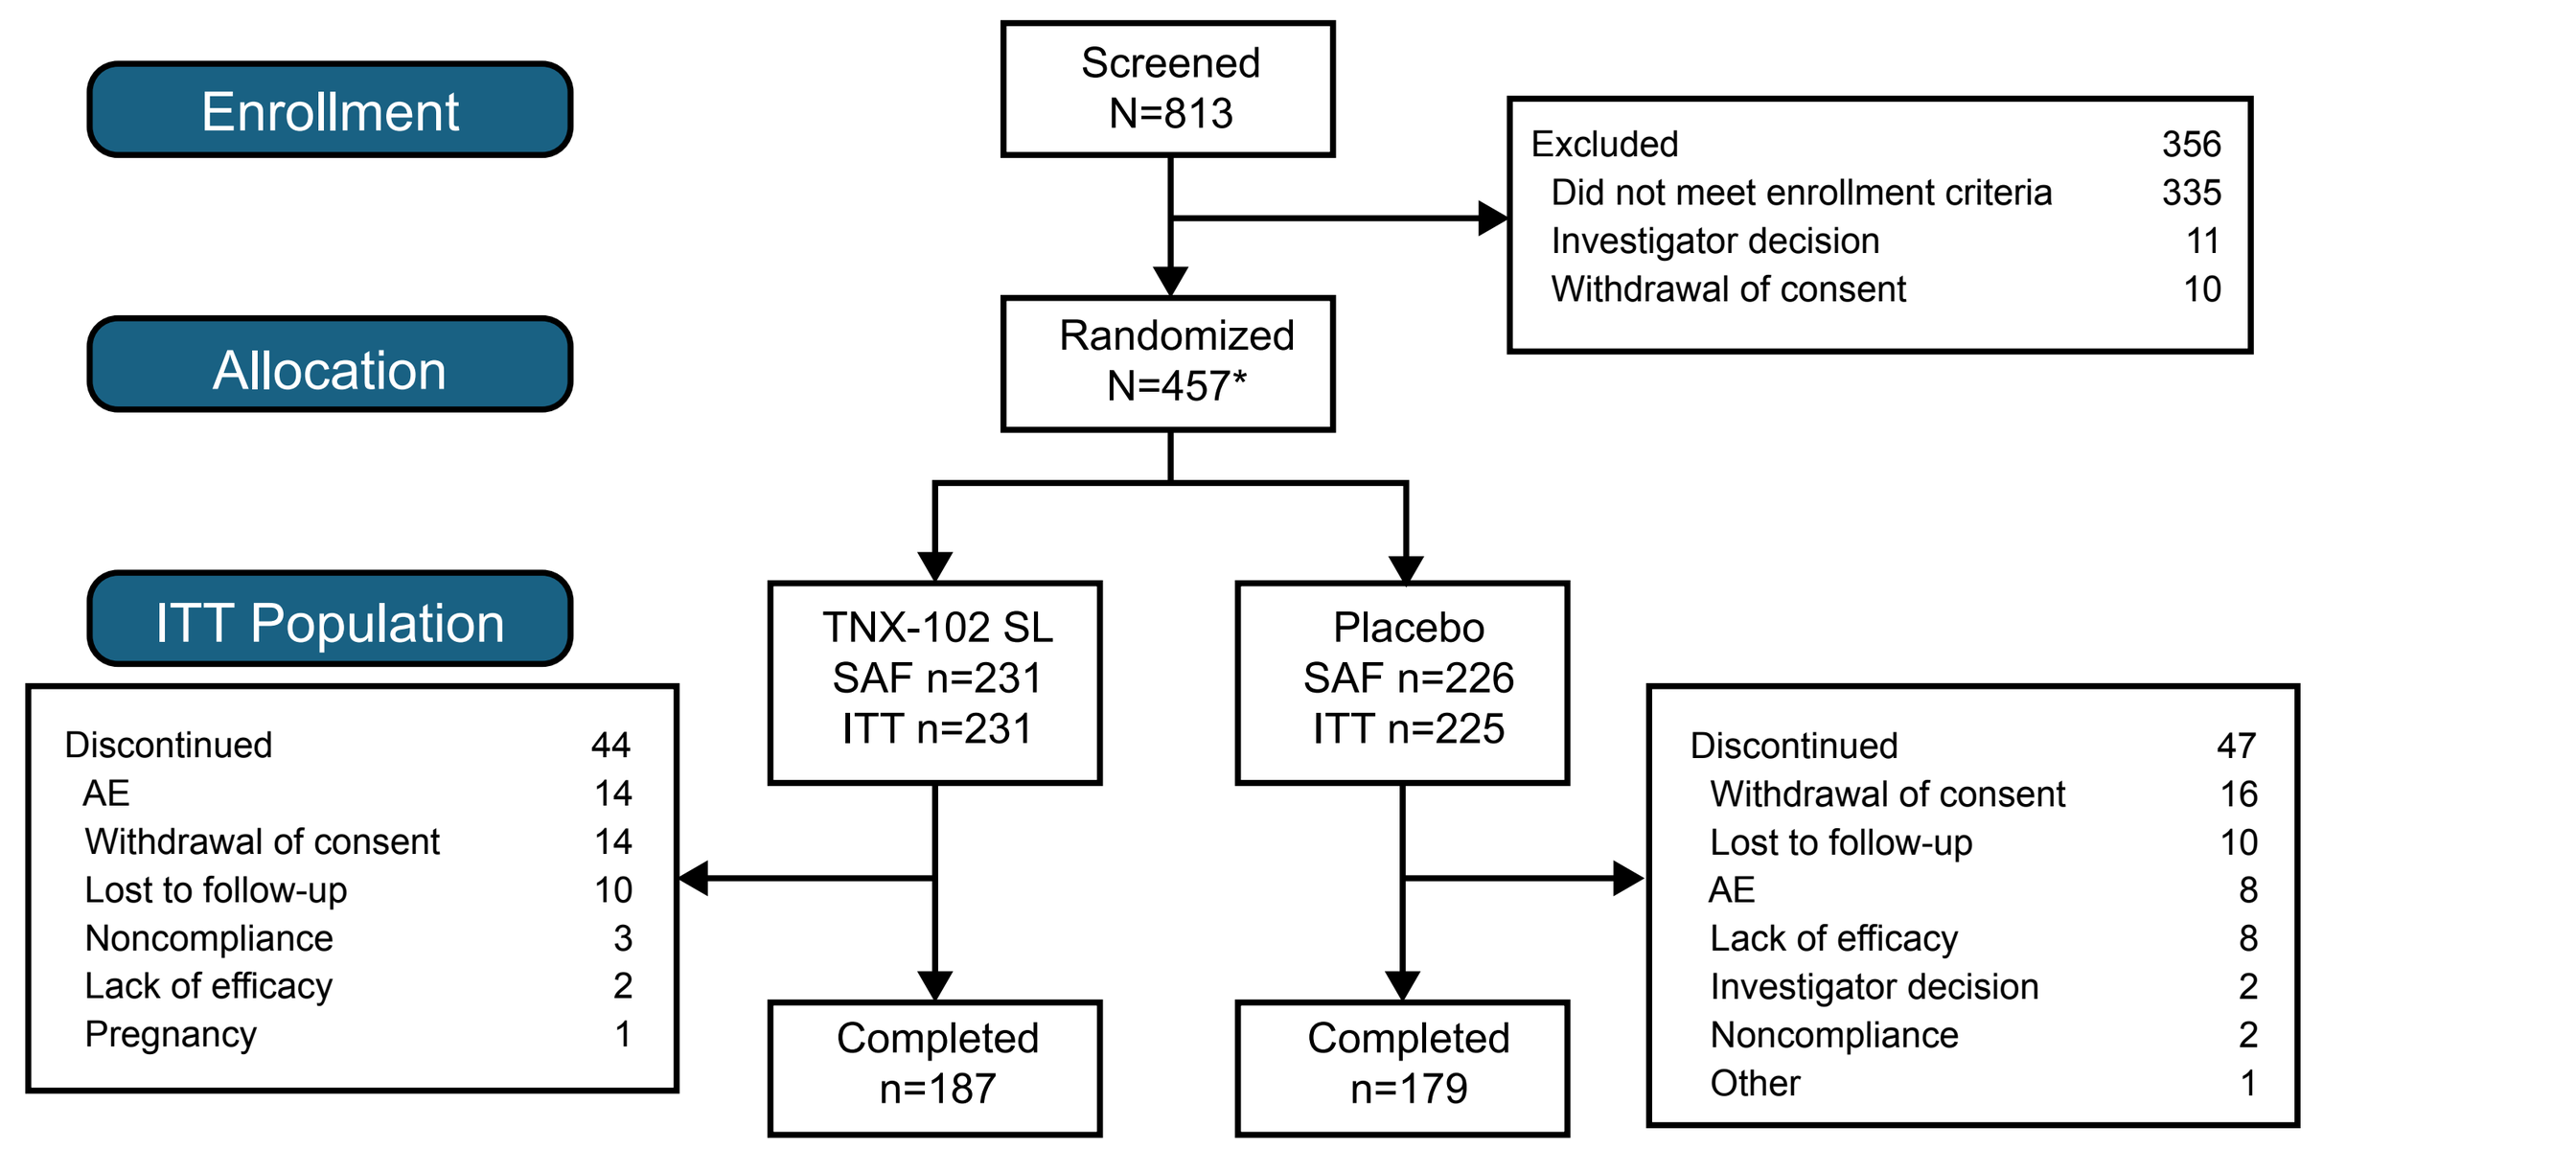
Enrollment
Allocation
ITT Population
Screened N=813
Excluded 356
Did not meet enrollment criteria 335
Investigator decision 11
Withdrawal of consent 10
Randomized N=457*
TNX-102 SL SAF n=231 ITT n=231
Placebo SAF n=226 ITT n=225
Discontinued 44
AE 14
Withdrawal of consent 14
Lost to follow-up 10
Noncompliance 3
Lack of efficacy 2
Pregnancy 1
Discontinued 47
Withdrawal of consent 16
Lost to follow-up 10
AE 8
Lack of efficacy 8
Investigator decision 2
Noncompliance 2
Other 1
Completed n=187
Completed n=179

*One placebo subject who was randomized in error (did not meet safety criteria) is included in Safety population (SAF) but not in ITT population.
AE, adverse event; ITT, intent-to-treat; SAF, safety.

Table 1: Participant Demographics and Baseline Characteristics (Safety Population)

	TNX-102 SL (n=231)	Placebo (n=226)	Total (N=457)
Age, years, mean (SD)	49.3 (10.5)	49.5 (11.3)	49.4 (10.9)
Female, n (%)	224 (97.0)	212 (93.8)	436 (95.4)
Race, n (%)			
White	194 (84.0)	192 (85.0)	386 (84.5)
Black/African American	32 (13.9)	27 (11.9)	59 (12.9)
Asian	1 (0.4)	5 (2.2)	6 (1.3)
Ethnicity, n (%)			
Hispanic or Latino	36 (15.6)	35 (15.5)	71 (15.5)
Weight, kg, mean (SD)	83.7 (17.8)	84.0 (17.4)	83.8 (17.8)
Employed currently, n (%)	147 (63.6)	150 (66.4)	297 (65.0)
Unable to work due to FM symptoms, n (%)	13 (5.6)	12 (5.3)	25 (5.5)
Education, some college or beyond, n (%)	187 (81.0)	193 (85.4)	380 (83.2)
Duration of FM disease in years, mean (SD)	8.6 (8.4)	9.9 (9.5)	9.2 (9.0)
Pain at baseline, NRS, mean (SD)*	5.9 (1.1)	5.9 (1.1)	—

*Pain reported for ITT population.
FM, fibromyalgia; ITT, intent to treat; NRS, numerical rating scale; SD, standard deviation.

Efficacy

- TNX-102 SL demonstrated significant improvement vs placebo (P<0.0001) in the primary endpoint of change in weekly pain scores at Week 14 (**Figure 2**)
 - At Week 14, the LS mean weekly average of daily pain scores was 4.1 (95% CI: 3.8, 4.3) for TNX-102 SL vs 4.7 (95% CI: 4.5, 5.0) for placebo
 - LS mean changes from baseline were -1.8 (95% CI: -2.0, -1.6) for TNX-102 SL vs -1.2 (95% CI: -1.4, -0.9) for placebo
 - The difference in LS means between TNX-102 SL and placebo was 0.7 (-1.0, -0.3) NRS units. The difference in the LS means (i.e., -0.7) was due to a rounding effect: the change from baseline was -1.82 in the TNX-102 group and -1.16 in the placebo group, and the difference in LS means between the groups was -0.66.

Figure 2: Mean (SE) Change From Baseline in Weekly Averages of Daily NRS Pain Scores

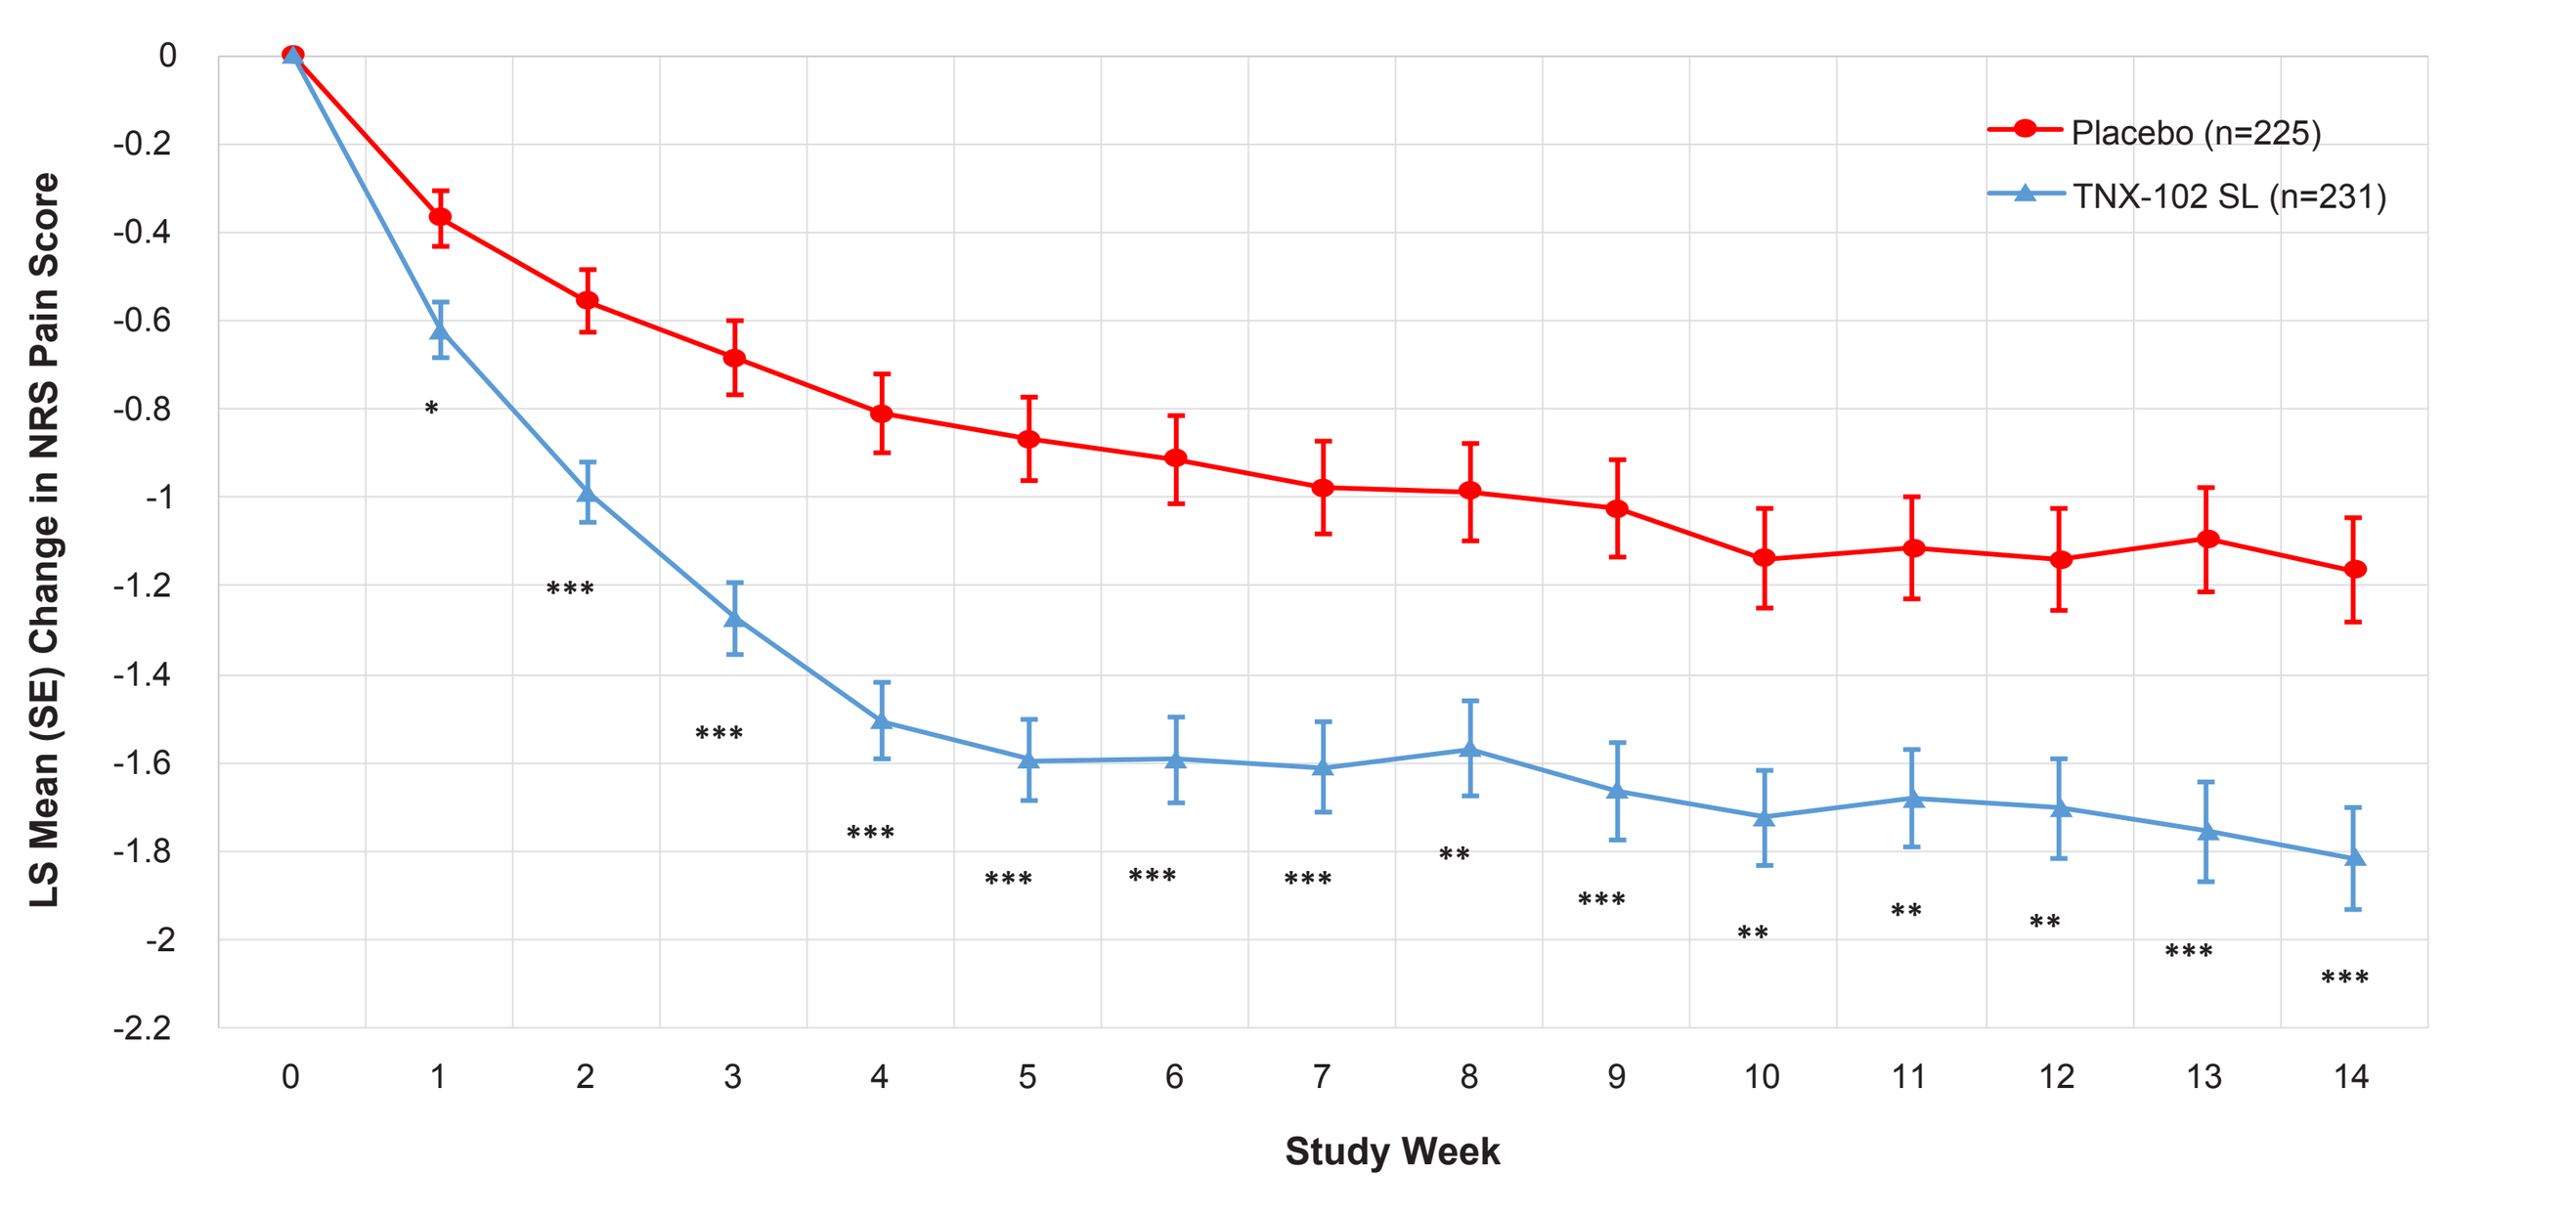
LS Mean (SE) Change in NRS Pain Score
Study Week
Placebo (n=225)
TNX-102 SL (n=231)
0
-0.2
-0.4
-0.6
-0.8
-1
-1.2
-1.4
-1.6
-1.8
-2
-2.2
0
1
2
3
4
5
6
7
8
9
10
11
12
13
14

*P<0.01; **P<0.001; ***P<0.0001. P-values for weeks 1-13 were not corrected for multiple comparisons.
LS, least-squares; NRS, numerical rating scale; SE, standard error.

- Female participants receiving TNX-102 SL showed larger mean improvements from baseline in total CSFQ-14 score at Week 14 vs those receiving placebo, reflecting improvements in sexual function (**Figure 3**).
 - Compared to placebo-treated participants, female participants treated with TNX-102 SL had similar or greater improvement in all domains. Treatment effects on desire/frequency (P=0.010), orgasm/completion (P=0.007), and total sexual function scores (P=0.010) were all nominally significant.
- The small number of male participants did not allow meaningful assessment of sexual functioning between treatment groups on the CSFQ-14

Figure 3: Mean (SE) Change From Baseline in CSFQ-14 Domains and Total Score in Females

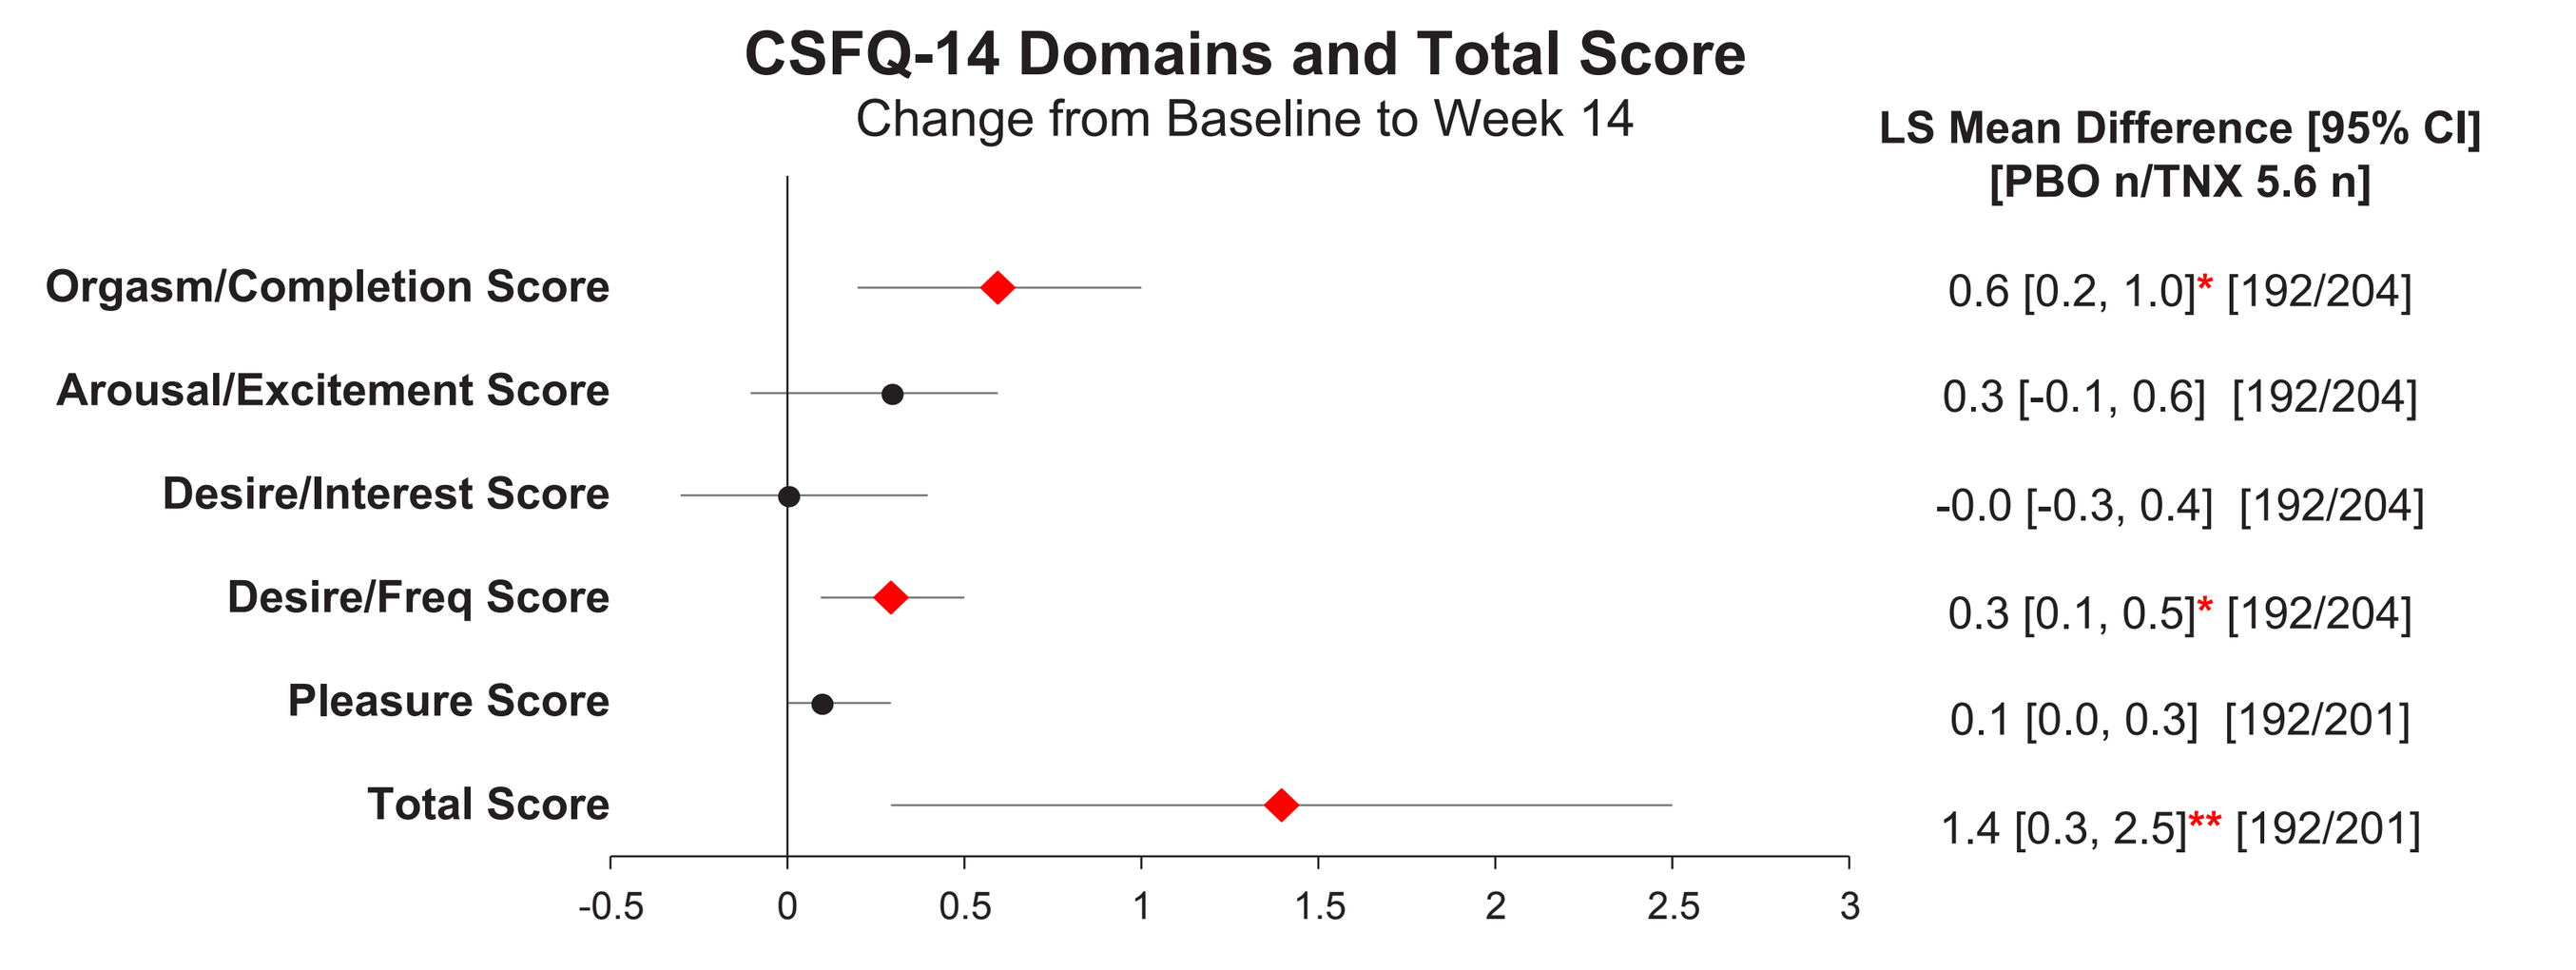
CSFQ-14 Domains and Total Score
Change from Baseline to Week 14
LS Mean Difference [95% CI]
[PBO n/TNX 5.6 n]
Orgasm/Completion Score
Arousal/Excitement Score
Desire/Interest Score
Desire/Freq Score
Pleasure Score
Total Score
0.6 [0.2, 1.0]* [192/204]
0.3 [-0.1, 0.6] [192/204]
-0.0 [-0.3, 0.4] [192/204]
0.3 [0.1, 0.5]* [192/204]
0.1 [0.0, 0.3] [192/201]
1.4 [0.3, 2.5]** [192/201]
-0.5
0
0.5
1
1.5
2
2.5
3

*P<0.05; **P<0.01.
Red diamonds indicate treatment differences with P<0.05; not corrected for multiple comparisons.
CI, confidence interval; CSFQ-14, Changes in Sexual Functioning Questionnaire short form; LS, least-squares; PBO, placebo; SE, standard error; TNX 5.6, TNX-102 SL 5.6 mg.

Safety

- No clinically meaningful differences in change from baseline in systolic or diastolic blood pressure or weight were observed between the TNX-102 SL and placebo groups at Week 14 (**Table 2**)

Table 2: Change From Baseline in Vital Sign Measurements

	TNX-102 SL (n=231)	Placebo (n=226)
Systolic blood pressure, mmHg		
n	185	180
Change from baseline to Week 14, mean (SD)	0.7 (12.4)	0.5 (10.4)
Diastolic blood pressure, mmHg		
n	185	180
Change from baseline to Week 14, mean (SD)	1.1 (8.6)	0.2 (8.2)
Weight, kg		
n	185	179
Change from baseline to Week 14, mean (SD)	0.02 (2.9)	0.20 (2.9)

- Treatment-emergent AEs (TEAEs) occurring in ≥3% of participants are shown in **Table 3**
 - A total of 47.9% of participants overall experienced at least one TEAE
 - Three (1.3%) participants each in both treatment groups experienced a severe TEAE. All other participants experiencing a TEAE rated the severity of their event as mild or moderate (57.6% of participants in the TNX-102 SL group and 35.4% of participants in the placebo group).
- The most commonly reported TEAEs were oral cavity administration site reactions that were mild-to-moderate, self-limited, and rarely led to discontinuations
 - Treatment-related discontinuations occurred in 6.1% and 3.5% of participants in the TNX-102 SL and placebo groups, respectively
 - Systemic TEAE rates, excluding COVID-19, remained below 4.0%

Table 3. TEAEs Occurring in ≥3% of Participants in Either Treatment Group

	TNX-102 SL (n=231)	Placebo (n=226)	Total (N=457)
Systemic adverse events, n (%)			
COVID-19	10 (4.3)	7 (3.1)	17 (3.7)
Somnolence	7 (3.0)	3 (1.3)	10 (2.2)
Headache	7 (3.0)	4 (1.8)	11 (2.4)
Oral cavity adverse events, n (%)			
Hypoesthesia oral	55 (23.8)	1 (0.4)	56 (12.3)
Product taste abnormal	27 (11.7)	2 (0.9)	29 (6.3)
Paresthesia oral	16 (6.9)	2 (0.9)	18 (3.9)
Tongue discomfort	16 (6.9)	0 (0.0)	16 (3.5)

CONCLUSIONS

- TNX-102 SL significantly reduced pain and showed a favorable tolerability profile, including minimal impact on weight and blood pressure
- Among females, TNX-102 SL was associated with improvements in sexual functioning compared with placebo and did not cause weight gain, an adverse effect often observed with other treatments
- Along with its unique mechanism targeting sleep disturbances, these results support TNX-102 SL as a new treatment option for FM
- The availability of a well-tolerated treatment may also encourage clinicians to make the diagnosis of FM earlier, thereby improving participant outcomes through timely intervention

References

1. Kang JH, et al. *J Rheum Dis.* 2022;29(1):4-13.
2. Theadom A, et al. *J Psychosom Res.* 2007;62(2):145-51.
3. Clauw DJ. *Ann Rheum Dis.* 2024;83(11):1421-7.
4. Kaplan CM, et al. *Nat Rev Neurol.* 2024;20(6):347-63.
5. Moldofsky H, et al. *Psychosom Med.* 1975;37(4):341-51.
6. Arout CA, et al. *J Womens Health (Larchmt).* 2018;27(8):1035-44.
7. Clauw DJ. *JAMA.* 2014;311(15):1547-55.
8. Robinson RL, et al. *Pain Med.* 2012;13(10):1366-76.
9. Lederman S, et al. *Pain Med.* 2025:pnaf089.
10. Lederman S, et al. *Arthritis Care Res (Hoboken).* 2023;75(11):2359-68.
11. Lederman S, et al. PainConnect 2025, April 3-6, 2025, Austin, TX.

Disclosures

GS, MK, JE, SE: Employee of and stock ownership in Tonix Pharmaceuticals, Inc.
EG: Employee of Tonix Medicines, Inc.

Contact info

Errol Gould, PhD, Tonix Medicines, Inc. 26 Main Street – Suite 101, Chatham, NJ 07928

Acknowledgements

This study was supported by Tonix Pharmaceuticals, Inc. (Chatham, NJ). Medical writing and editorial assistance were provided by Nicole L. Day, PhD, MWC, and Anthony DiLauro, PhD, ELS, of the Sensified Division of Woven Health Collective, LLC (New York NY), and were funded by Tonix Pharmaceuticals, Inc.

Presented at the American College of Rheumatology Convergence Conference 2025, October 24-29, 2025, Chicago, IL